Cannabidiol (CBD) Significantly Reduces Convulsive Seizure Frequency in Dravet Syndrome (DS): Results of a Multi-center, Randomized, Double-blind, Placebo-controlled Trial (GWPCARE1)

J. Helen Cross[1] | Orrin Devinsky[2] | Linda Laux[3] | Eric Marsh[4] | Ian Miller[5] | Rima Nabbout[6] | Ingrid E Scheffer[7] | Elizabeth A. Thiele[8] | Stephen Wright[9]

[1]UCL Great Ormond Street Institute of Child Health, London, UK; [2]NYU Comprehensive Epilepsy Center, New York, NY; [3]Ann and Robert H Lurie Children's Hospital of Chicago, Lurie Children's Epilepsy Center, Chicago, Illinois; [4]The Children's Hospital of Philadelphia, Philadelphia PA; [5]Miami Children's Hospital, Miami, Florida; [6]Hôpital Necker - Enfants Malades, Paris, France; [7]University of Melbourne, Florey Institute, Austin Health and Royal Children's Hospital, Melbourne, Australia; [8]Massachusetts General Hospital, Boston, MA; [9]GW Research, Ltd, London, England.

SUMMARY

- The trial met its primary endpoint, demonstrating that CBD (20 mg/kg/day), as an add-on to standard of care, produced significantly greater reductions in convulsive seizures vs. placebo in patients with Dravet syndrome (DS).
- CBD resulted in significantly greater reductions in total seizure frequency vs. placebo.
- Compared to placebo, CBD caregivers were significantly more likely to report an improvement in overall condition, as measured on the Caregiver Global Impression of Change (CGIC) scale.
- CBD resulted in more adverse events than placebo, but it was generally well tolerated.

INTRODUCTION

- Data from a US expanded access program suggest that CBD reduces seizures in patients with DS[1].
- This is the first randomized, double-blind, placebo-controlled trial to investigate the efficacy and safety of adjunctive CBD in children (2–18 years old) with DS.

Patient disposition and baseline demographics

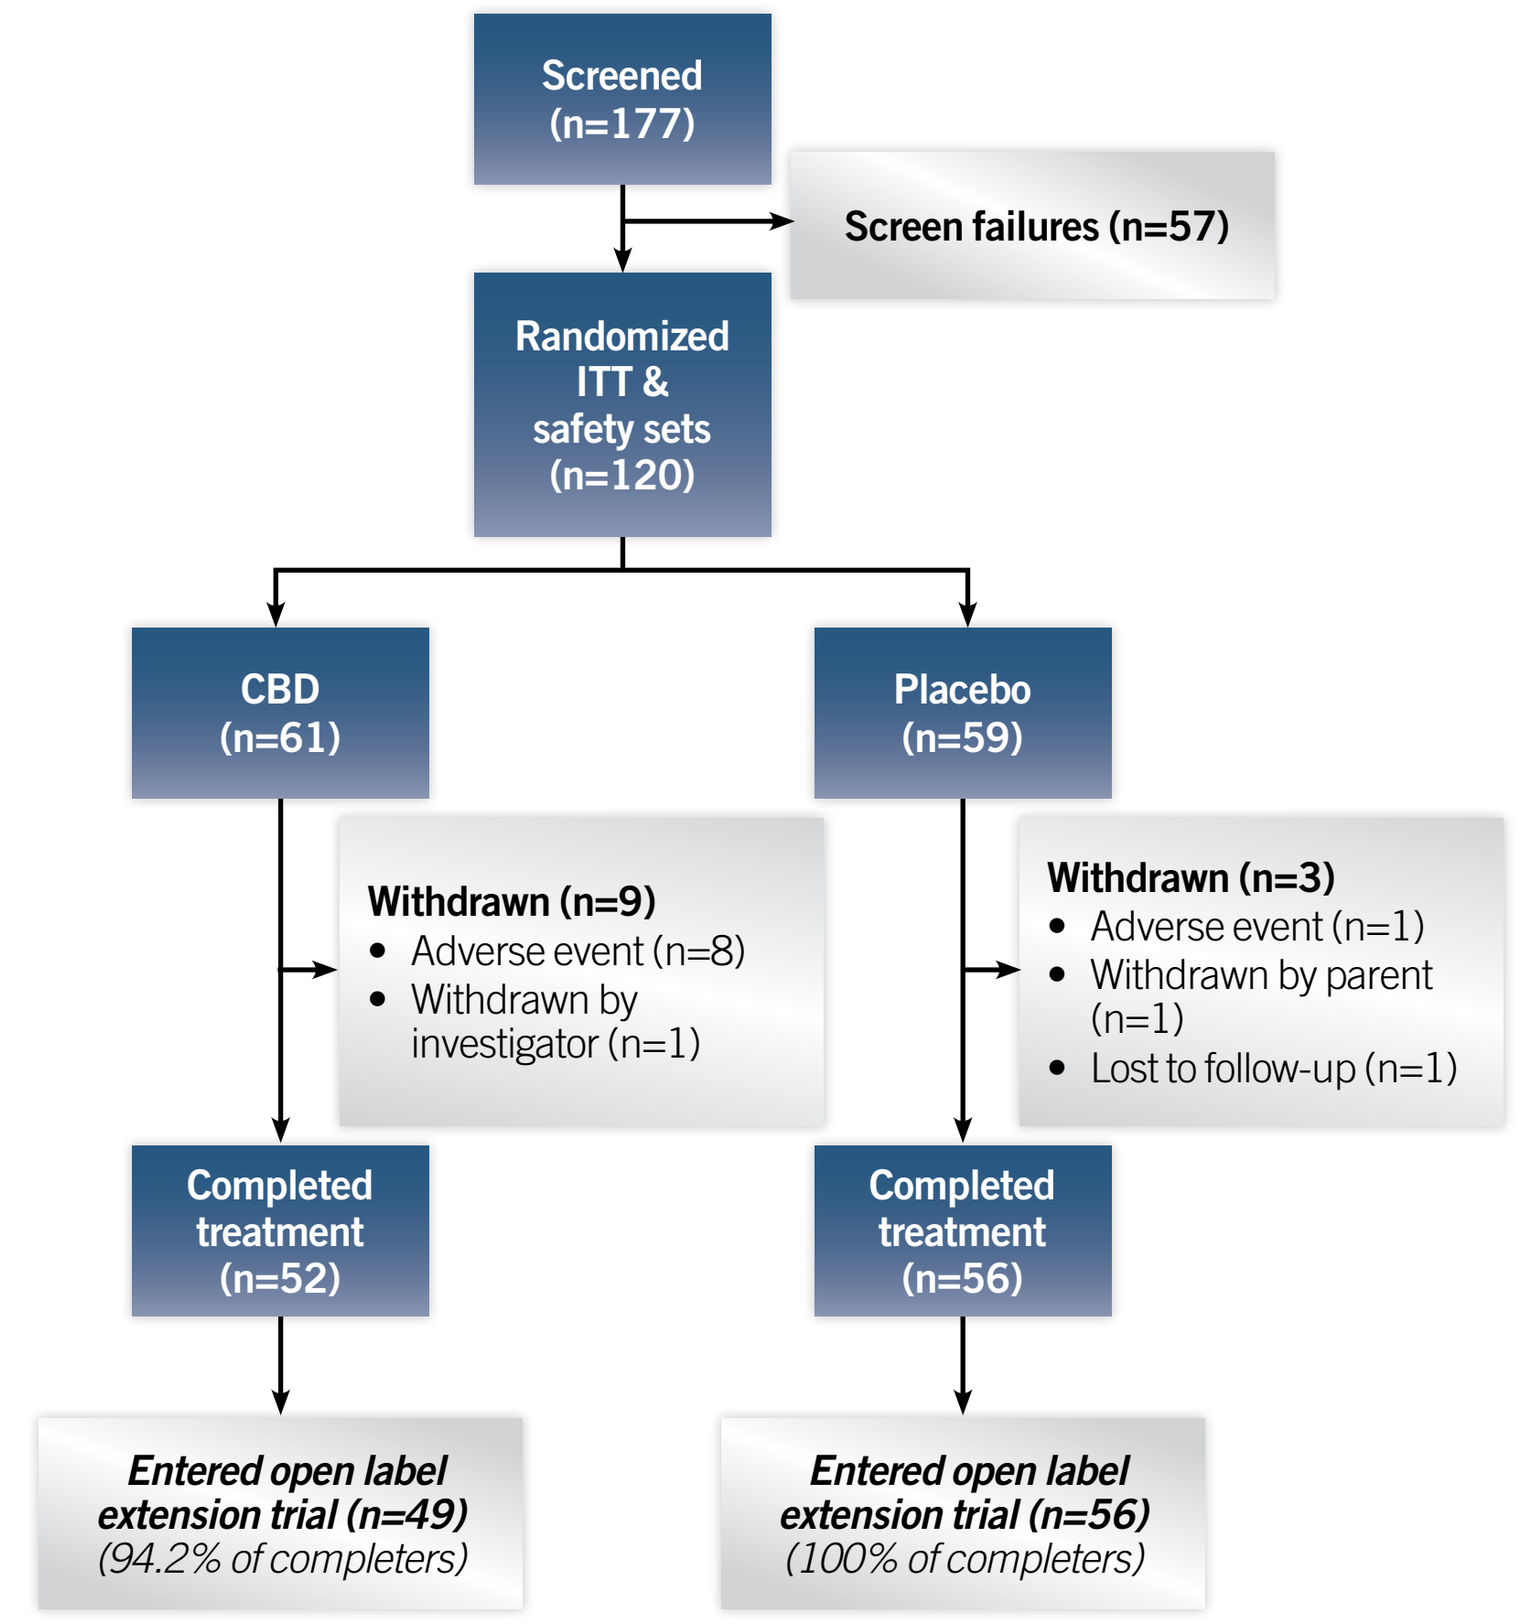
Screened (n=177)
Screen failures (n=57)
Randomized ITT & safety sets (n=120)
CBD (n=61)
Placebo (n=59)
Withdrawn (n=9)
• Adverse event (n=8)
• Withdrawn by investigator (n=1)
Withdrawn (n=3)
• Adverse event (n=1)
• Withdrawn by parent (n=1)
• Lost to follow-up (n=1)
Completed treatment (n=52)
Completed treatment (n=56)
Entered open label extension trial (n=49) (94.2% of completers)
Entered open label extension trial (n=56) (100% of completers)

Parameter	CBD (n=61)	Placebo (n=59)
Age (years)		
Mean (min, max)	9.7 (2.5, 18.0)	9.8 (2.3, 18.4)
Age Group [n (%)]		
2–5 years	18 (29.5)	17 (28.8)
6–12 years	23 (37.7)	24 (40.7)
13–18 years	20 (32.8)	18 (30.5)
Sex [n (%)]		
Male	35 (57.4)	27 (45.8)
Number of Prior AEDs		
Median (min, max)	4 (0, 26)	4 (0, 14)
Number of Current AEDs*		
Median (min, max)	3 (1, 5)	3 (1, 5)
Seizure Frequency (per 28 Days) During Baseline		
Median Convulsive (Q1, Q3)	12.4 (6.2, 28.0)	14.9 (7.0, 36.0)
Median Total (Q1, Q3)	24 (10.4, 141.0)	41.5 (12.0, 367.0)

*Most common antiepileptic drugs (AEDs) were clobazam (65%), valproic acid (57%), stiripentol (43%).

EFFICACY RESULTS

Percentage change in convulsive and total seizure frequency (28-day average)

A | Convulsive Seizures

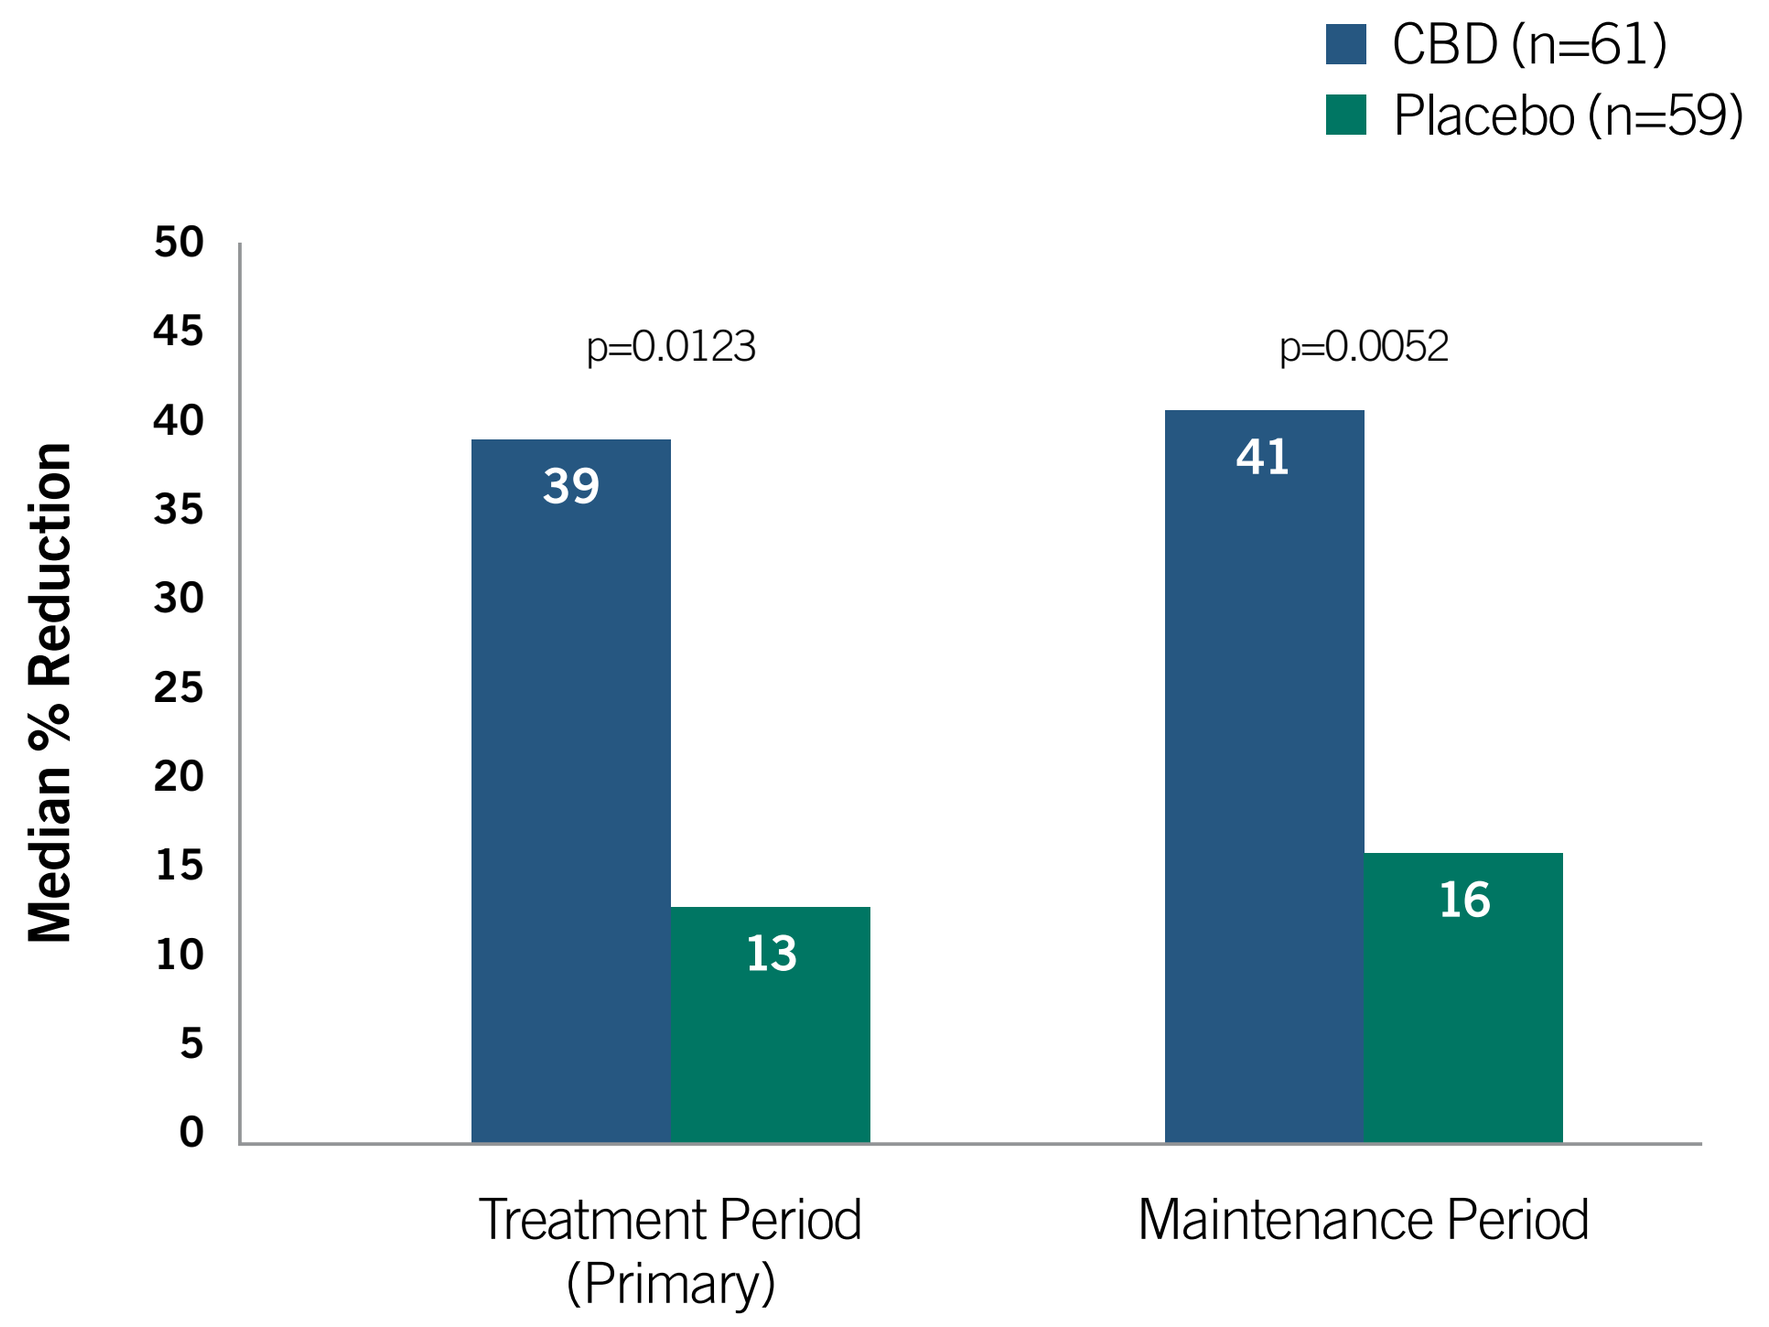
CBD (n=61)
Placebo (n=59)
Median % Reduction
p=0.0123
p=0.0052
39
13
41
16
Treatment Period (Primary)
Maintenance Period

B | Total Seizures

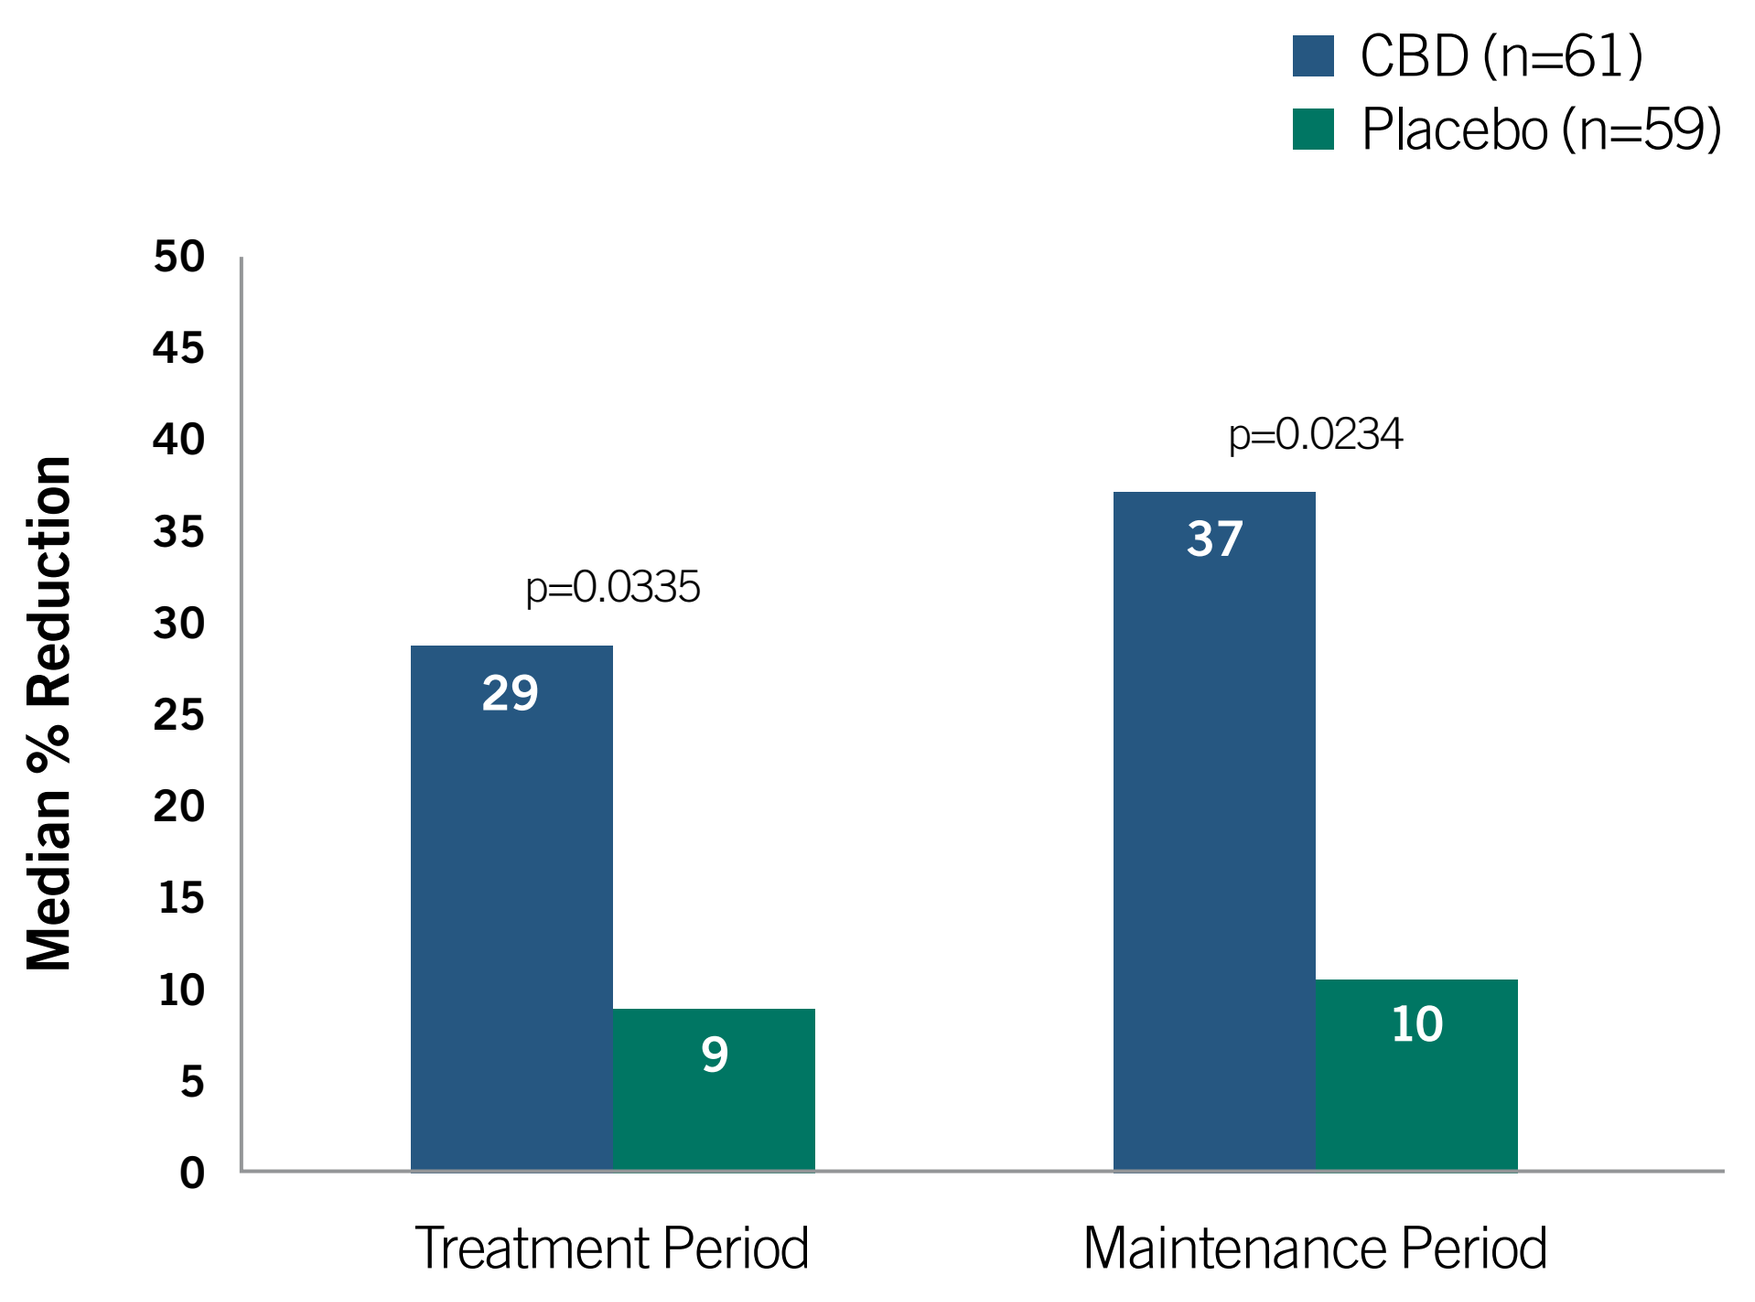
CBD (n=61)
Placebo (n=59)
Median % Reduction
p=0.0335
p=0.0234
29
9
37
10
Treatment Period
Maintenance Period

- Significantly greater reductions were reported for CBD than for placebo; this difference was established during the first 4 weeks of maintenance.
- Reduction in non-convulsive seizures was not statistically different for subgroup of patients with non-convulsive seizures who were taking CBD (n=37, 40%) vs. placebo (n=41, 35%).

Convulsive seizure responder rates

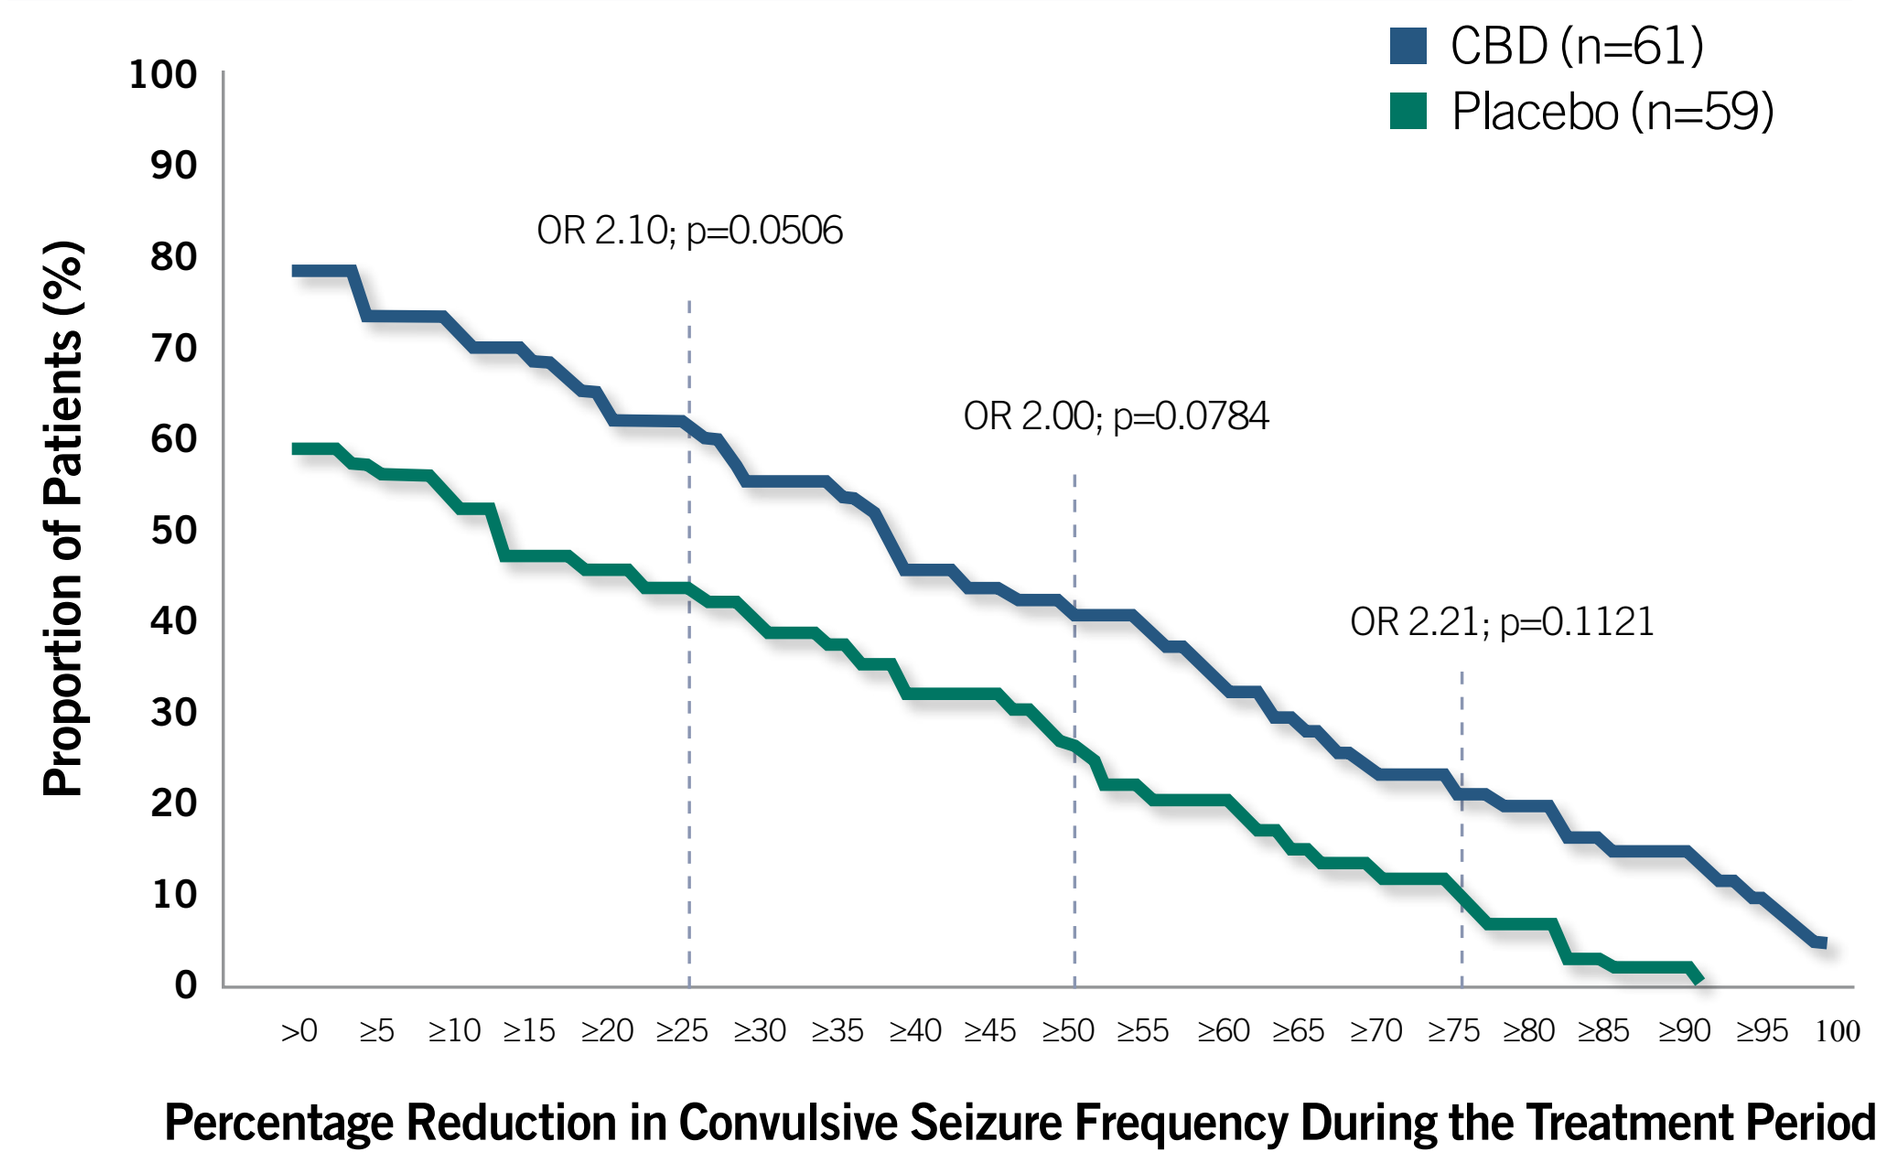
CBD (n=61)
Placebo (n=59)
Proportion of Patients (%)
OR 2.10; p=0.0506
OR 2.00; p=0.0784
OR 2.21; p=0.1121
>0 ≥5 ≥10 ≥15 ≥20 ≥25 ≥30 ≥35 ≥40 ≥45 ≥50 ≥55 ≥60 ≥65 ≥70 ≥75 ≥80 ≥85 ≥90 ≥95 100
Percentage Reduction in Convulsive Seizure Frequency During the Treatment Period

Caregiver global impression of change from baseline at last visit

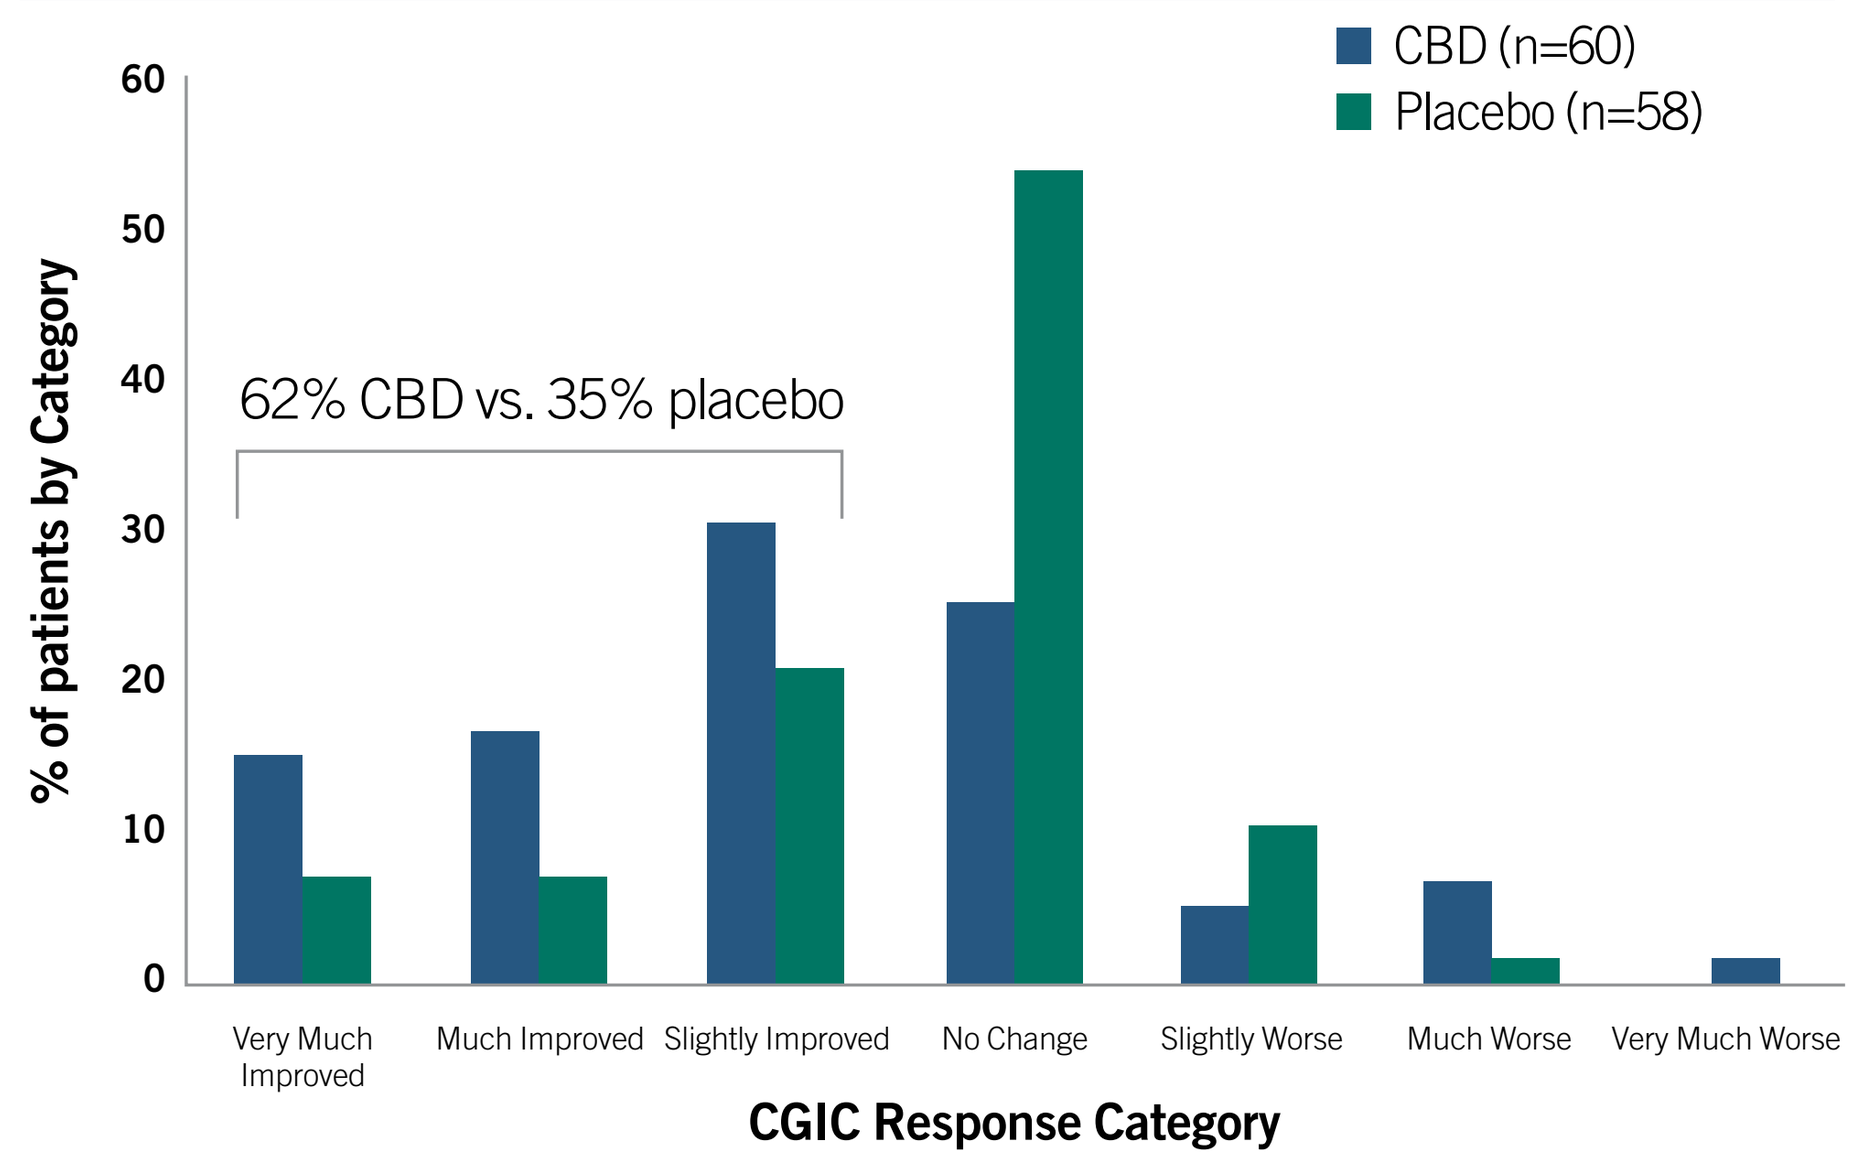
CBD (n=60)
Placebo (n=58)
% of patients by Category
62% CBD vs. 35% placebo
Very Much Improved
Much Improved
Slightly Improved
No Change
Slightly Worse
Much Worse
Very Much Worse
CGIC Response Category

- 43% of CBD patients had a ≥50% reduction in convulsive seizures compared to 27% of patients taking placebo.
- 3 CBD and no placebo patients achieved convulsive and total seizure freedom during the entire treatment period. An additional 4 CBD patients achieved convulsive seizure freedom during maintenance (no placebo); of these, 1 completed the trial.
- Caregivers of CBD patients were more likely to report an improvement in overall condition on CGIC (OR=2.29; p=0.0155).
- 93% of patients were on multiple concomitant AEDs. The effect of concomitant AEDs on efficacy will be explored in future pooled analyses.

SAFETY RESULTS

Summary of treatment-emergent adverse events (TEAEs)

	CBD (n=61) n (%)	Placebo (n=59) n (%)
All-causality TEAEs	57 (93.4%)	44 (74.6%)
Treatment-related TEAEs	43 (70.5%)	16 (27.1%)
TEAEs leading to withdrawal	9 (14.8%)	1 (1.7%)
Serious TEAEs	10 (16.4%)	3 (5.1%)
Treatment-related serious TEAEs	5 (8.2%)	0
TEAEs reported in >10% of patients in either group by preferred term		
Somnolence	22 (36.1%)	6 (10.2%)
Diarrhea	19 (31.1%)	6 (10.2%)
Decreased appetite	17 (27.9%)	3 (5.1%)
Fatigue	12 (19.7%)	2 (3.4%)
Pyrexia	9 (14.8%)	5 (8.5%)
Vomiting	9 (14.8%)	3 (5.1%)
Lethargy	8 (13.1%)	3 (5.1%)
Upper respiratory tract infection	7 (11.5%)	5 (8.5%)
Convulsion	7 (11.5%)	3 (5.1%)

- Of those who reported a TEAE, 84% of CBD and 95% of placebo patients reported it as mild or moderate.
- The pattern of serious TEAEs was consistent with the common TEAEs reported on CBD.
- There was no difference in the number of patients who experienced status epilepticus between CBD (n=4) and placebo (n=3); no episodes were deemed treatment-related nor led to withdrawal from treatment.
- There were no deaths.

Laboratory investigations

- Increases in ALT or AST (>3× ULN) occurred in 12 CBD and 1 placebo patient, all of whom were on concomitant valproic acid. No patients met standard criteria for drug-induced liver injury (Hy's law) with concurrent elevated bilirubin >2× ULN. Three of the CBD patients withdrew from treatment. All elevations resolved.

METHODS

- Eligible patients were aged 2–18 years, with a clinical diagnosis of DS inadequately controlled by ≥1 current AED(s), and were experiencing ≥4 convulsive seizures during the 28-day baseline period.
- Patients were randomized (1:1) to 20 mg/kg/day of a pharmaceutical formulation of CBD (100 mg/mL) in oral solution or matched placebo, administered b.i.d. starting at 2.5 mg/kg/day and titrated up to 20 mg/kg/day over a 2-week period, followed by a 12-week dose maintenance period.
- Convulsive seizures were defined as tonic–clonic, tonic, clonic, and atonic seizures.
- The treatment period consisted of both the titration and maintenance periods.
- Caregivers recorded seizures daily using an automated interactive voice response system.
- DS diagnosis and classification of seizure types was confirmed by the Epilepsy Study Consortium.
- Patients who completed the trial were eligible to continue into an open-label extension study.

Disclosures: This study was sponsored by GW Pharmaceuticals plc, London, UK. Formatting and editorial assistance was provided to the authors by Hospicom, Cold Spring, NY and funded by GW Pharmaceuticals. All authors met the ICMJE authorship criteria. Neither honoraria nor payments were made for authorship. J. Helen Cross, Orrin Devinsky, Linda Laux, Eric Marsh, Ian Miller, Rima Nabbout, Ingrid E Scheffer, and Elizabeth A. Thiele have consulted for, conducted studies funded by, or received honoraria from GW Pharmaceuticals, plc. Stephen Wright is an employee of GW Research, Ltd. Findings reported in this study are specific to GW Pharmaceuticals' formulation of cannabidiol and cannot be extrapolated to other cannabidiol products.

References: 1. Devinsky O, Marsh E, Friedman D, et al. Cannabidiol in patients with treatment-resistant epilepsy: an open-label interventional trial. *Lancet Neurol.* 2015;4422(15):1–9.

Contact Information: medinfo.usa@gwpharm.com

Presented: The American Epilepsy Society Annual Meeting; Houston, TX; December 2-6, 2016.

To obtain a PDF of this poster
- Scan the QR code or
- Visit www.GWQRcodes.com/311151

Charges may apply. No personal information is stored.

GREENWICH™ BIOSCIENCES
A GW Pharmaceuticals PLC Company